Enhanced bioavailability and reduced absorption variability of Oral PTH 1-34 in men

Gregory Burshtein [1], Jonathan C. Y. Tang[2], Hillel Galitzer [1], Ariel Rothner [1], Phillip Schwartz [1], William D. Fraser2, Yoseph Caraco[3]

[1]Entera Bio Ltd., Israel, [2] Bioanalytical Facility, Biomedical Research Centre, Norwich Medical School, Faculty of Medicine and Health Sciences, University of East Anglia, Norwich, UK, [3] Hadassah Clinical Research Center, Israel

BACKGROUND

The oral absorption of polypeptides, is characterized by high dose-to-dose variability due to their low bioavailability. As a result, maintaining the blood drug levels within the therapeutic window is very challenging.

An orally administered PTH may have prodigious advantages in the treatment of bone related disorders due to improved patient compliance and adherence. However, in order for PTH, a drug whose specific PK profile is critical for its physiological activity, to be effective and safe, a consistent and reproducible absorption profile is essential.

Entera Bio previously developed and presented an oral formulation of PTH(1-34) that achieved biologically relevant plasma concentrations of the drug similar to those of the commercial SC injection. In anticipation of entering more advanced clinical studies, further development of oral PTH(1-34) formulation was performed focusing on the control of the drug absorption while minimizing its variability. The main source of high variability was revealed through numerous *in vitro* and preclinical studies resulting in the development of a novel and improved oral PTH (1-34) formulations.

We now present results from a clinical study utilizing the novel formulation technology in our oral PTH (1-34) tablets with decreased inter-subject variability and significantly increased bioavailability.

STUDY DESIGN

A Phase I, open label, crossover design pharmacokinetic - pharmacodynamic study was conducted at the Hadassah Clinical Research Center at the Hebrew University – Hadassah Medical Center. Nine healthy male volunteers received Entera Bio's original oral formulation (OF) and two modified oral formulations (MF-1 and MF-2) at the fixed dose of 1.5 mg PTH (1-34). Blood samples were analyzed externally at the Bioanalytical Facility at the University of East Anglia by validated chemiluminescence based assay on the IDS-ISYS automated analyzer. Levels of serum calcium were measured at the clinical laboratory of the Hadassah hospital.

RESULTS

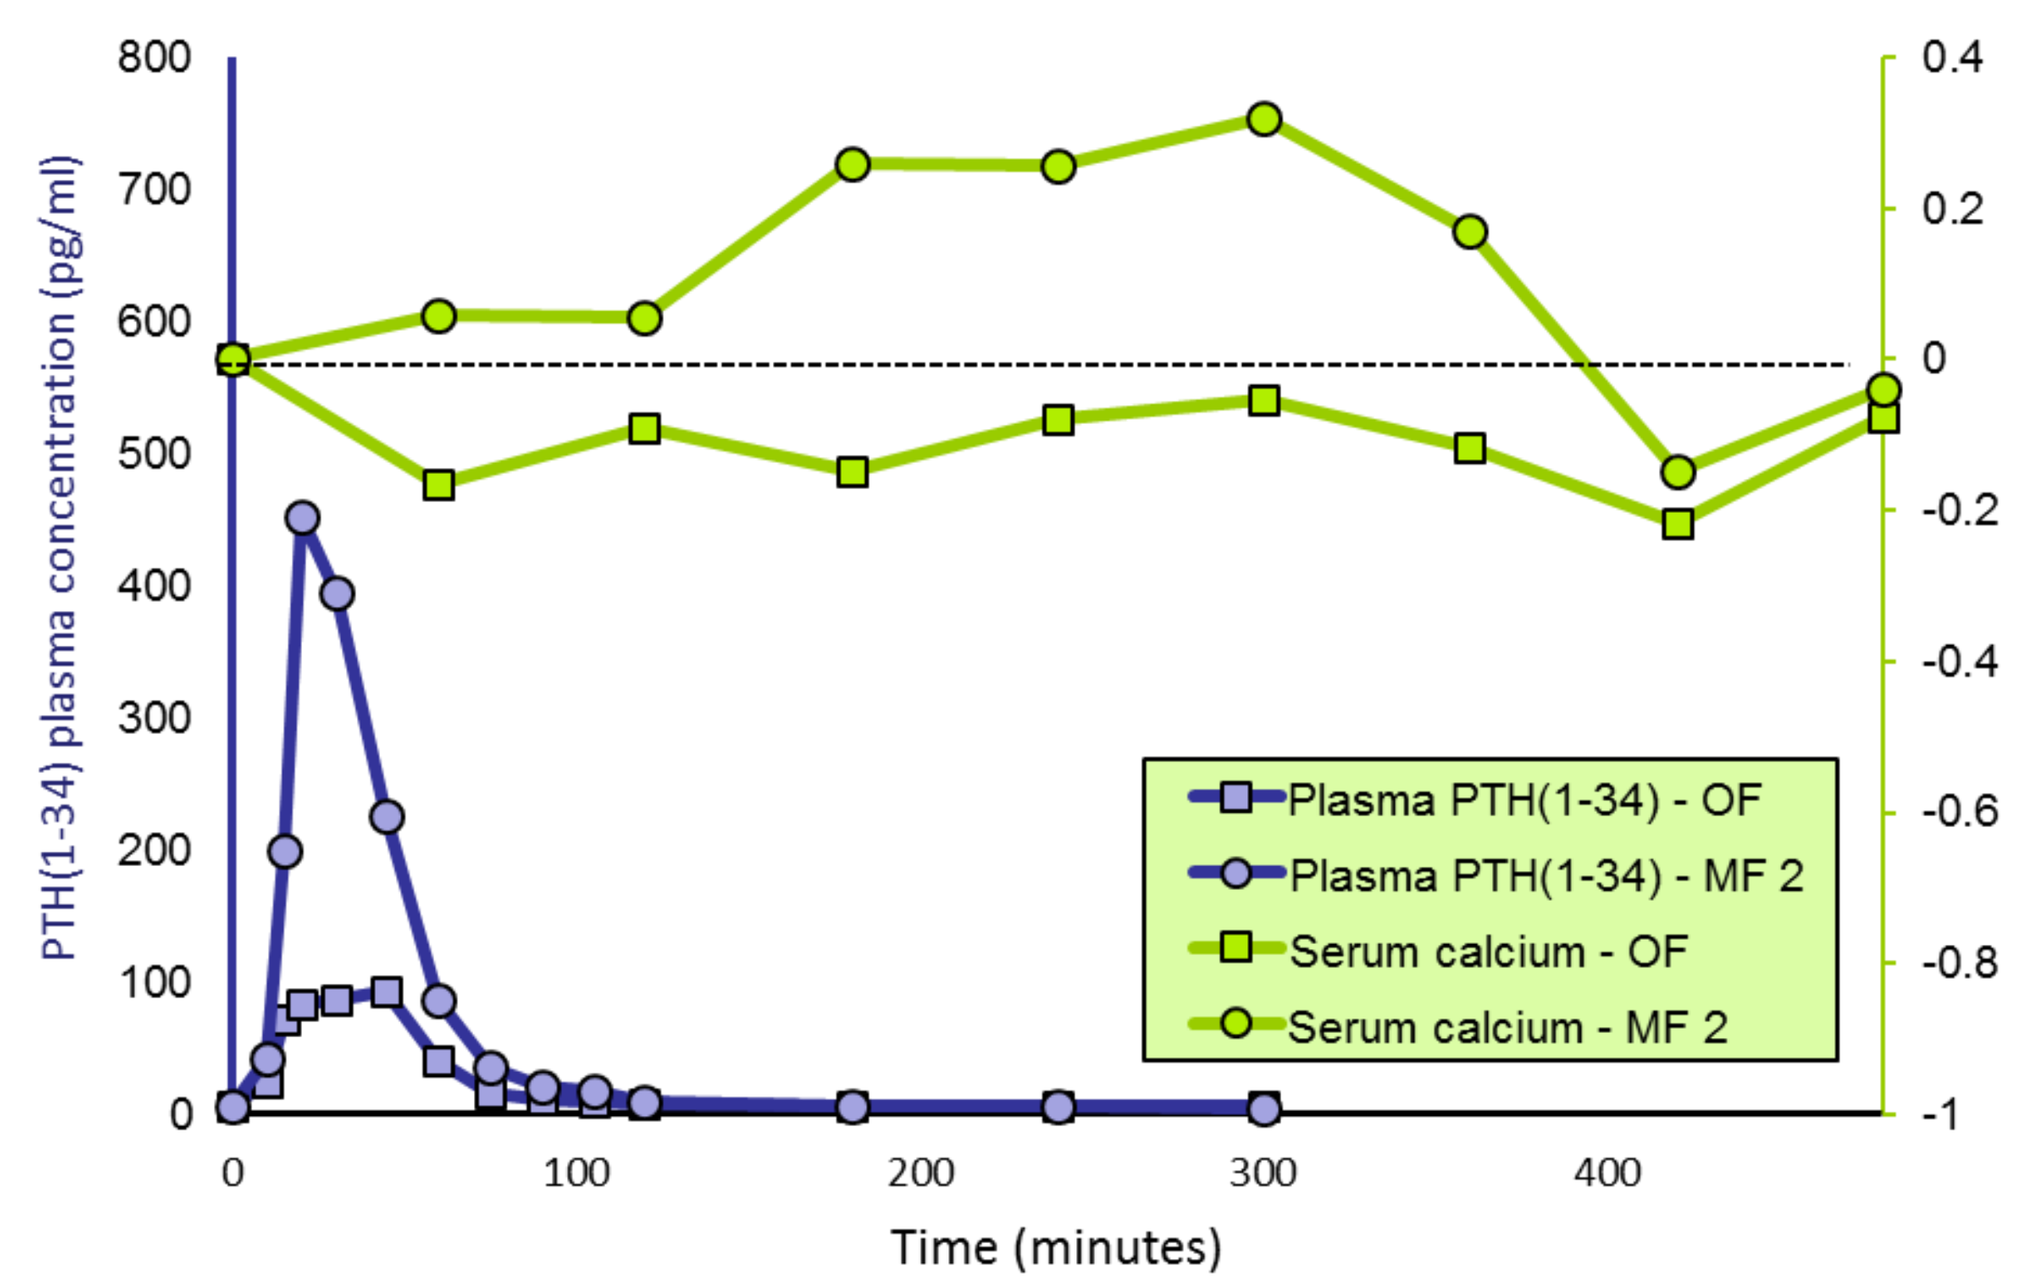
PTH(1-34) plasma concentration (pg/ml)
Serum albumin adjusted Calcium (mg/dL)
Time (minutes)
Plasma PTH(1-34) - OF
Plasma PTH(1-34) - MF 2
Serum calcium - OF
Serum calcium - MF 2

Figure 1. ***PK*** *and PD profiles following the administration of original and second modified oral formulation of PTH (1-34) 1.5mg (as base). Pharmacodynamic profile expressed as a change in the levels of albumin adjusted serum calcium. Each data point represents the mean of 9 volunteers.*

The pharmacokinetic (PK) profile of all oral PTH (1-34) formulations is characterized by rapid absorption and elimination rates of the drug.

The systemic exposure (AUC) and maximal plasma concentrations (C_{max}) following the administration of the modified oral PTH(1-34) formulations (MF-1 and MF-2) were noticably higher than AUC and C_{max} of original formulation (OF) (Table 1). C_{max} and AUC of the MF-2 were statistically significantly higher than C_{max} and AUC of the original formulation ($p = 0.005$ and $p = 0.01$, respectively). C_{max} coefficients of variation (CV%) of the basic oral formulation, modified formulation 1 and modified formulation 2 were 123%, 91% and 67%, respectively (Table 1; Figure 2).

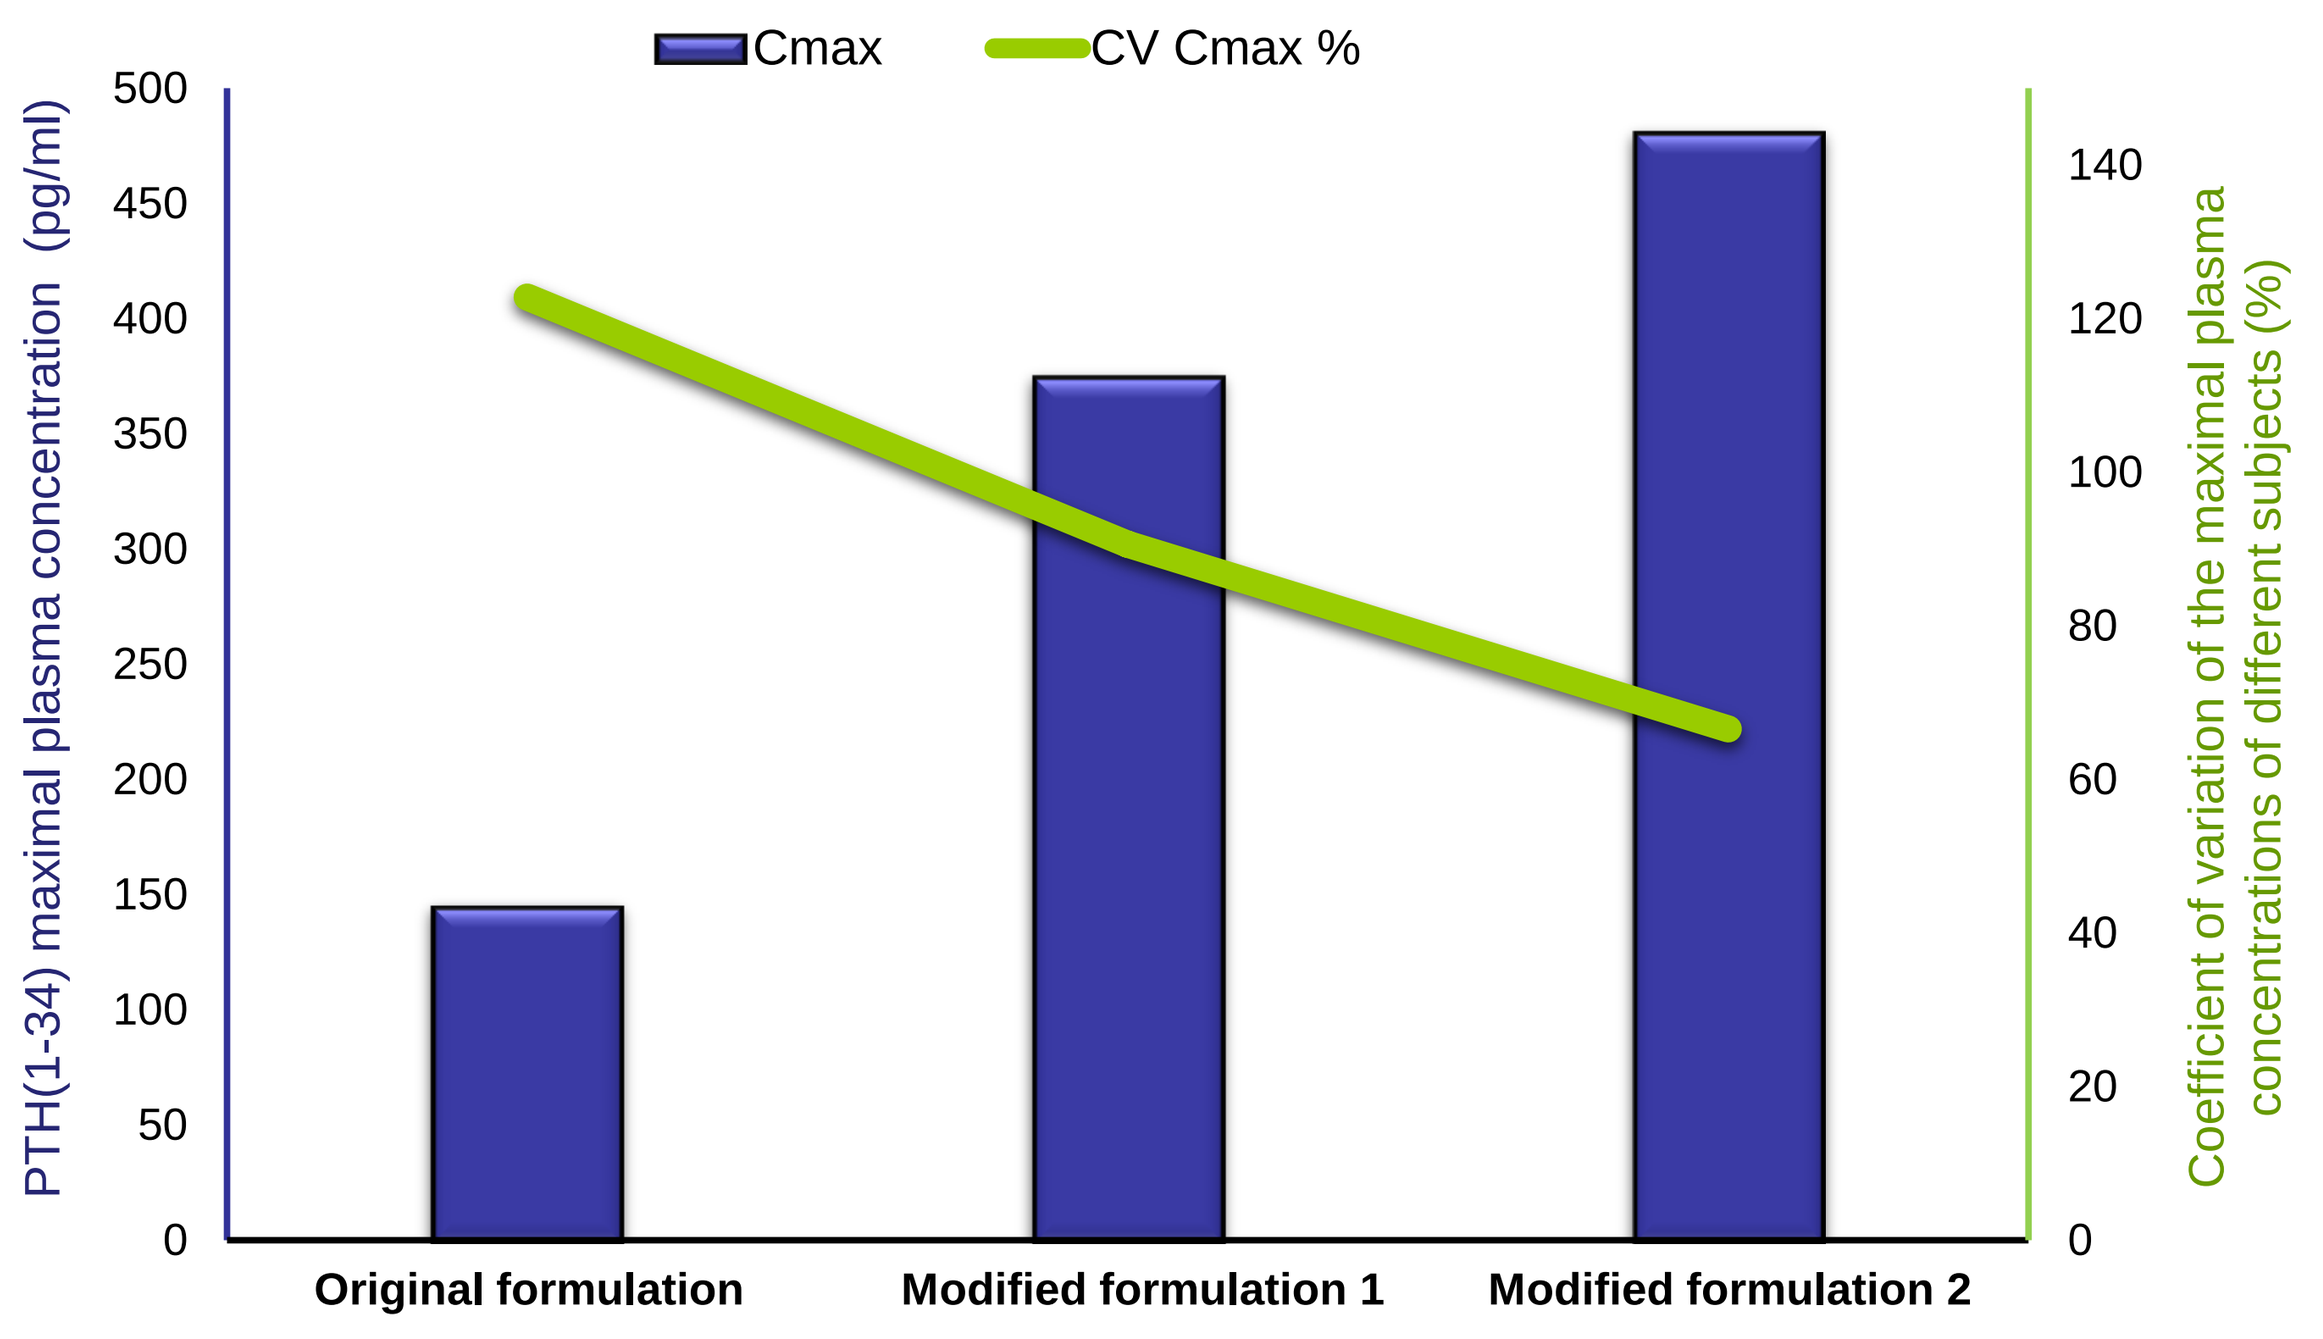
Cmax
CV Cmax %
PTH(1-34) maximal plasma concentration (pg/ml)
Coefficient of variation of the maximal plasma concentrations of different subjects (%)
Original formulation
Modified formulation 1
Modified formulation 2

Figure 2. *Maximal PTH (1-34) plasma concentrations and its coefficient of variation (C_{max} CV%) following administration of OF, MF-1 and MF-2, fixed dose (1.5 mg) oral formulations of PTH (1-34). Each data point represents the mean of 9 healthy volunteers.*

Similarly, to the drug absorption, pharmacodynamic (PD) response of the MF-2, presented as the maximal relative increase in albumin adjusted calcium, in comparison to the OF was improved from 0.07±0.29mg/dL to 0.32±0.24mg/dL (Table 1; Figure 1).

RESULTS

PTH (1-34) formulation	n	C_{max} (pg/ml)	C_{max} CV (%)	AUC (pg*min/ml)	Serum Calcium increase (mg/dL)
Original oral formulation (OF)	9	145.1 ± 56	123	3481.2 ± 1843	0.07 ± 0.33
Modified oral formulation 1 (MF-1)	9	374.9 ± 108	91	7976 ± 2556	0.12 ± 0.21
Modified oral formulation 2 (MF-2)	9	480.8 ± 101*	67	11369.4 ± 3719*	0.32 ± 0.33

Table 1. *Summary of the main pharmacokinetic and pharmacodynamic parameters found in the current study. C_{max} – the maximal PTH (1-34) plasma concentration; C_{max} CV% - coefficient of variation among the C_{max} levels of different volunteers; AUC - total drug exposure; Serum Calcium increase – change in albumin corrected serum Calcium relatively to the baseline; All the data presented as a mean ± SE. * statistically significant in comparison to the original oral formulation of PTH (1-34) ($p \leq 0.05$).*

CONCLUSIONS

Modification of the original Entera Bio oral delivery system of PTH (1-34) resulted in significantly greater bioavailability and reduced variability in both the C_{max} values and the total drug exposure, AUC.

Presented results focused on C_{max} which is the most relevant factor for determining biological activation of anabolic pathways and calcium regulation. In the specific case of Entera Bio's oral PTH (1-34) delivery system, this essential 'sharp and short' exposure to the drug is achieved due to the fast absorption of the molecule and its rapid elimination from the body. Similarly, the drug pharmacodynamic effect was also enhanced by the novel oral formulation of PTH(1-34).

Reduced inter-subject C_{max} variability, enhanced bioavailability and an increased pharmacodynamic effect achieved by the modification of the dosage form, ensure an improved safety profile of oral PTH (1-34) on the one hand and biologically effective blood concentrations of the drug on the other hand. These together with the expected improvement in compliance significantly enhance the potential of Entera Bio's oral PTH(1-34) becoming a clinical success.

Hadassah Ein-Kerem, Jerusalem Bio Park, POB 12117 Jerusalem 9112002 Israel ©Copyright Entera Bio Ltd.

Printed by Call4Posters